A DNA plasmid melanoma cancer vaccine, SCIB1, combined with nivolumab + ipilimumab in patients with advanced unresectable melanoma: Efficacy and safety results from the open-label Phase 2 SCOPE Trial

Heather Shaw[1,2], Poulam Patel[3], Miranda Payne[4], Satish Kumar[5], Sarah Danson[6], Martin Highley[7], Clare Barlow[8], Kellati Prasad[9], Ioannis Karydis[10], Maria Marples[11], Kate Young[12], Pippa Corrie[13], Robert Miller, Georgia Goodhew, Samantha Paston, Gaelle Cane, Joe Chadwick, Lindy Durrant and Nermeen Varawalla[14]

1- Mount Vernon Cancer Centre, Hertfordshire, 2- University College London Hospital, London, 3 - Nottingham Hospitals University Trust, Nottingham, 4- Churchill Hospital, Oxford University Hospitals, Oxford, 5- Velindre Cancer Centre, Velindre University NHS Trust, Cardiff, Wales, 6- Weston Park Cancer Centre, Sheffield Teaching Hospital NHS Trust, Sheffield, 7- Derriford Hospital, University Hospitals NHS Trust, Plymouth, 8- Musgrove Park Hospital, Somerset NHS Foundation Trust, Taunton, 9- Royal Preston Hospital, Preston, 10- Southampton General Hospital, Southampton, 11- St James's University Hospital, Leeds Teaching Hospitals NHS Trust, Leeds, 12- The Royal Marsden Hospital, London, 13- Addenbrooke's Hospital, Cambridge, 14- Scancell Ltd, Oxford, United Kingdom

SCANCELL

ABSTRACT

BACKGROUND

Targeting melanoma with T cells drives anti-tumor responses. We previously showed that a DNA vaccine, SCIB1, incorporating T cell epitopes from TRP-2/gp100 into an antibody framework to allow Fc targeting of activated dendritic cells, was successfully a monotherapy in Stage 3/4 melanoma patients in a Phase 1/2 clinical study. Unresectable melanoma patients showed a 60% ORR and 88% of patients treated with SCIB1, post tumor resection, remained disease free for 5 years [1]. The current SCOPE trial tests the hypothesis that unresectable patients may have an improved response when SCIB1 is combined with checkpoint inhibitors (CPI) compared to CPI alone.

METHODS

Patients are treated with nivolumab with ipilimumab and SCIB1 (8mg) i.m. using needle-free injection at a fixed dosing schedule for up to 10 doses over 24 months. The CPI therapy was administered i.v. in accordance with their respective SmPC. ORR as measured by RECIST 1.1 in the overall intention-to-treat population was the primary endpoint. The study is designed using Simon's two stage methodology with 80% power when the true response rate is 70% versus 50% RR of no interest with an overall type I error of 5%. In the first stage, 15 patients needed to be enrolled and if there are eight or fewer clinical responses (RECIST 1.1 objective response [CR or PR] within 25 weeks of the first dose of SCIB1), further recruitment would have been stopped. The null hypothesis will be rejected if 27 or more responses are observed in 43 patients. T cell responses were assessed using a cultured ELISpot assay following TRP-2 or gp100 peptides stimulation

RESULTS

To date, 43 patients received the combination of SCIB1 with nivolumab and ipilimumab. 25 patients had reached 25 weeks, and had 80% PFS, 84% DCR and 72% ORR with 20% of patients showing a complete response. 94% (16/17) of the clinical responders had a T cell response, 80% (4/5) of the patients with stable disease had a T cell response, 33% (1/3) of patients with progressive disease had a T cell response. T cell receptors isolated from patients showed strong, peptide, ImmunoBody® and tumor recognition. Of interest was that in patient who showed a pre-vaccinated response to the encoded epitope a different TCR was stimulated by the vaccine. Most of the SCIB1-related adverse events were Grade 1/2. Four Grade 3 related adverse events have been reported each in one patient: rash, neutropenia, pulmonary embolism and raised GGT. No exacerbation of immune-mediated adverse events was observed when SCIB1 was added to nivolumab with ipilimumab.

CONCLUSIONS

SCIB1 in combination with nivolumab and ipilimumab as first line treatment for unresectable Stage 3/4 melanoma resulted in 80% 6-month PFS with strong vaccine specific T cell responses without an increase in clinically meaningful adverse events. These results, if confirmed, in a larger patient cohort provide confidence in initiating a randomized registration program in unresectable melanoma patients with our novel DNA plasmid technology.

SCIB1

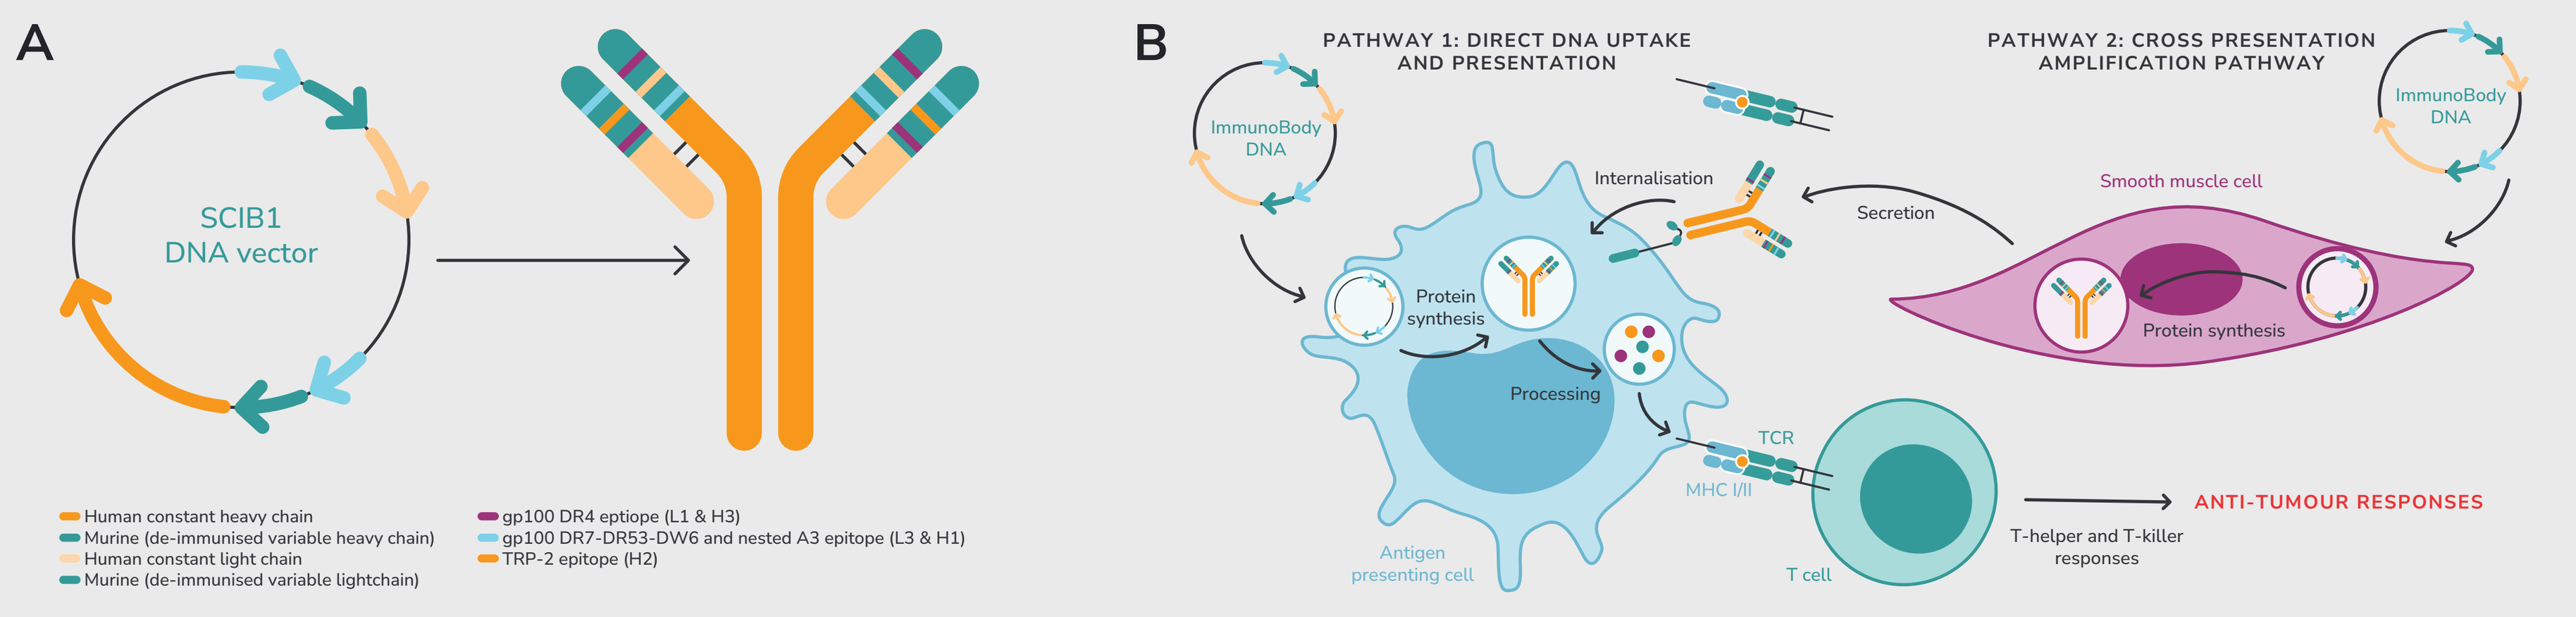
A
SCIB1 DNA vector
Human constant heavy chain
Murine (de-immunised variable heavy chain)
Human constant light chain
Murine (de-immunised variable lightchain)
gp100 DR4 epitope (L1 & H3)
gp100 DR7-DR53-DW6 and nested A3 epitope (L3 & H1)
TRP-2 epitope (H2)
B
PATHWAY 1: DIRECT DNA UPTAKE AND PRESENTATION
PATHWAY 2: CROSS PRESENTATION AMPLIFICATION PATHWAY
ImmunoBody DNA
Internalisation
Secretion
Smooth muscle cell
Protein synthesis
Processing
TCR
MHC I/II
Antigen presenting cell
T cell
ANTI-TUMOUR RESPONSES
T-helper and T-killer responses

(A) SCIB1, an off-the-shelf DNA vaccine incorporates CD8 and CD4 T cell epitopes from melanoma antigens TRP-2 and gp100 in an APC targeting antibody framework (B) SCIB1 elicits anti-tumour immune responses by both direct presentation, uptake of plasmid and expression of engineered antibody by antigen-presenting cells & cross presentation, secretion of the engineered antibody targeted to CD64 FcγR present on dendritic cells via its Fc domain.

SCOPE PHASE 2 CLINICAL TRIAL DESIGN

Phase 2 open label study to evaluate safety and efficacy of SCIB1 administered in combination with check point inhibitors Primary objective to determine in the intention-to-treat population, the ORR, as measured by RECIST 1.1.

43 patients with advanced unresectable melanoma with target HLA haplotype, namely, positive for A2 and at least one of DR4, DR7, DR53 or DQ6, enrolled at 15 clinical trial sites in the United Kingdom.

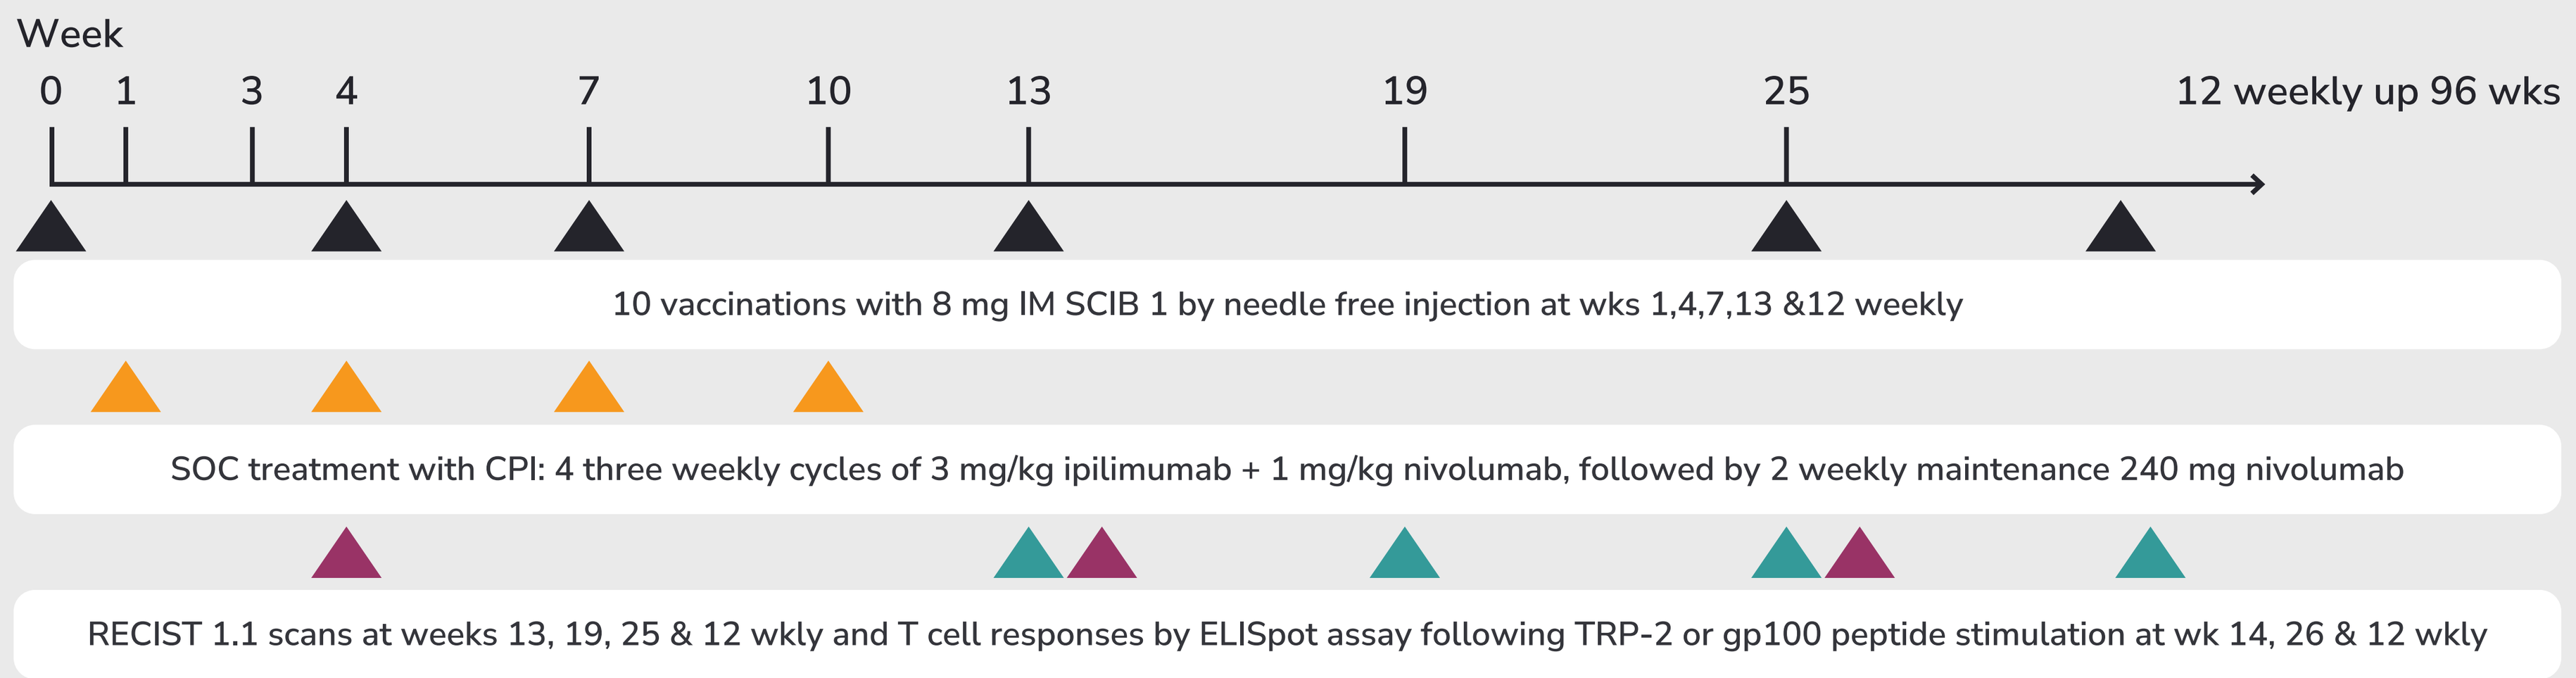
Week
0
1
3
4
7
10
13
19
25
12 weekly up 96 wks
10 vaccinations with 8 mg IM SCIB 1 by needle free injection at wks 1,4,7,13 &12 weekly
SOC treatment with CPI: 4 three weekly cycles of 3 mg/kg ipilimumab + 1 mg/kg nivolumab, followed by 2 weekly maintenance 240 mg nivolumab
RECIST 1.1 scans at weeks 13, 19, 25 & 12 wkly and T cell responses by ELISpot assay following TRP-2 or gp100 peptide stimulation at wk 14, 26 & 12 wkly

PRELIMINARY RESULTS

The results of the first 25 immunized patients who have completed their 25-week scan or experienced disease progression are presented here. The study seeks to evaluate improvement in ORR relative to 50.6% reported for 1 mg/kg nivolumab plus 3mg/kg ipilimumab {2}. As per the Simon Stage Design, at stage 1, 8 or fewer clinical responses (CR+PR) within 25 weeks of first immunization amongst the first 15 patients enrolled would call for recruitment to be stopped for futility. This stage was successfully passed and now the study has fully recruited 43 patients. At Simon stage 2, the null hypothesis would be rejected if there are 27 or more responses.

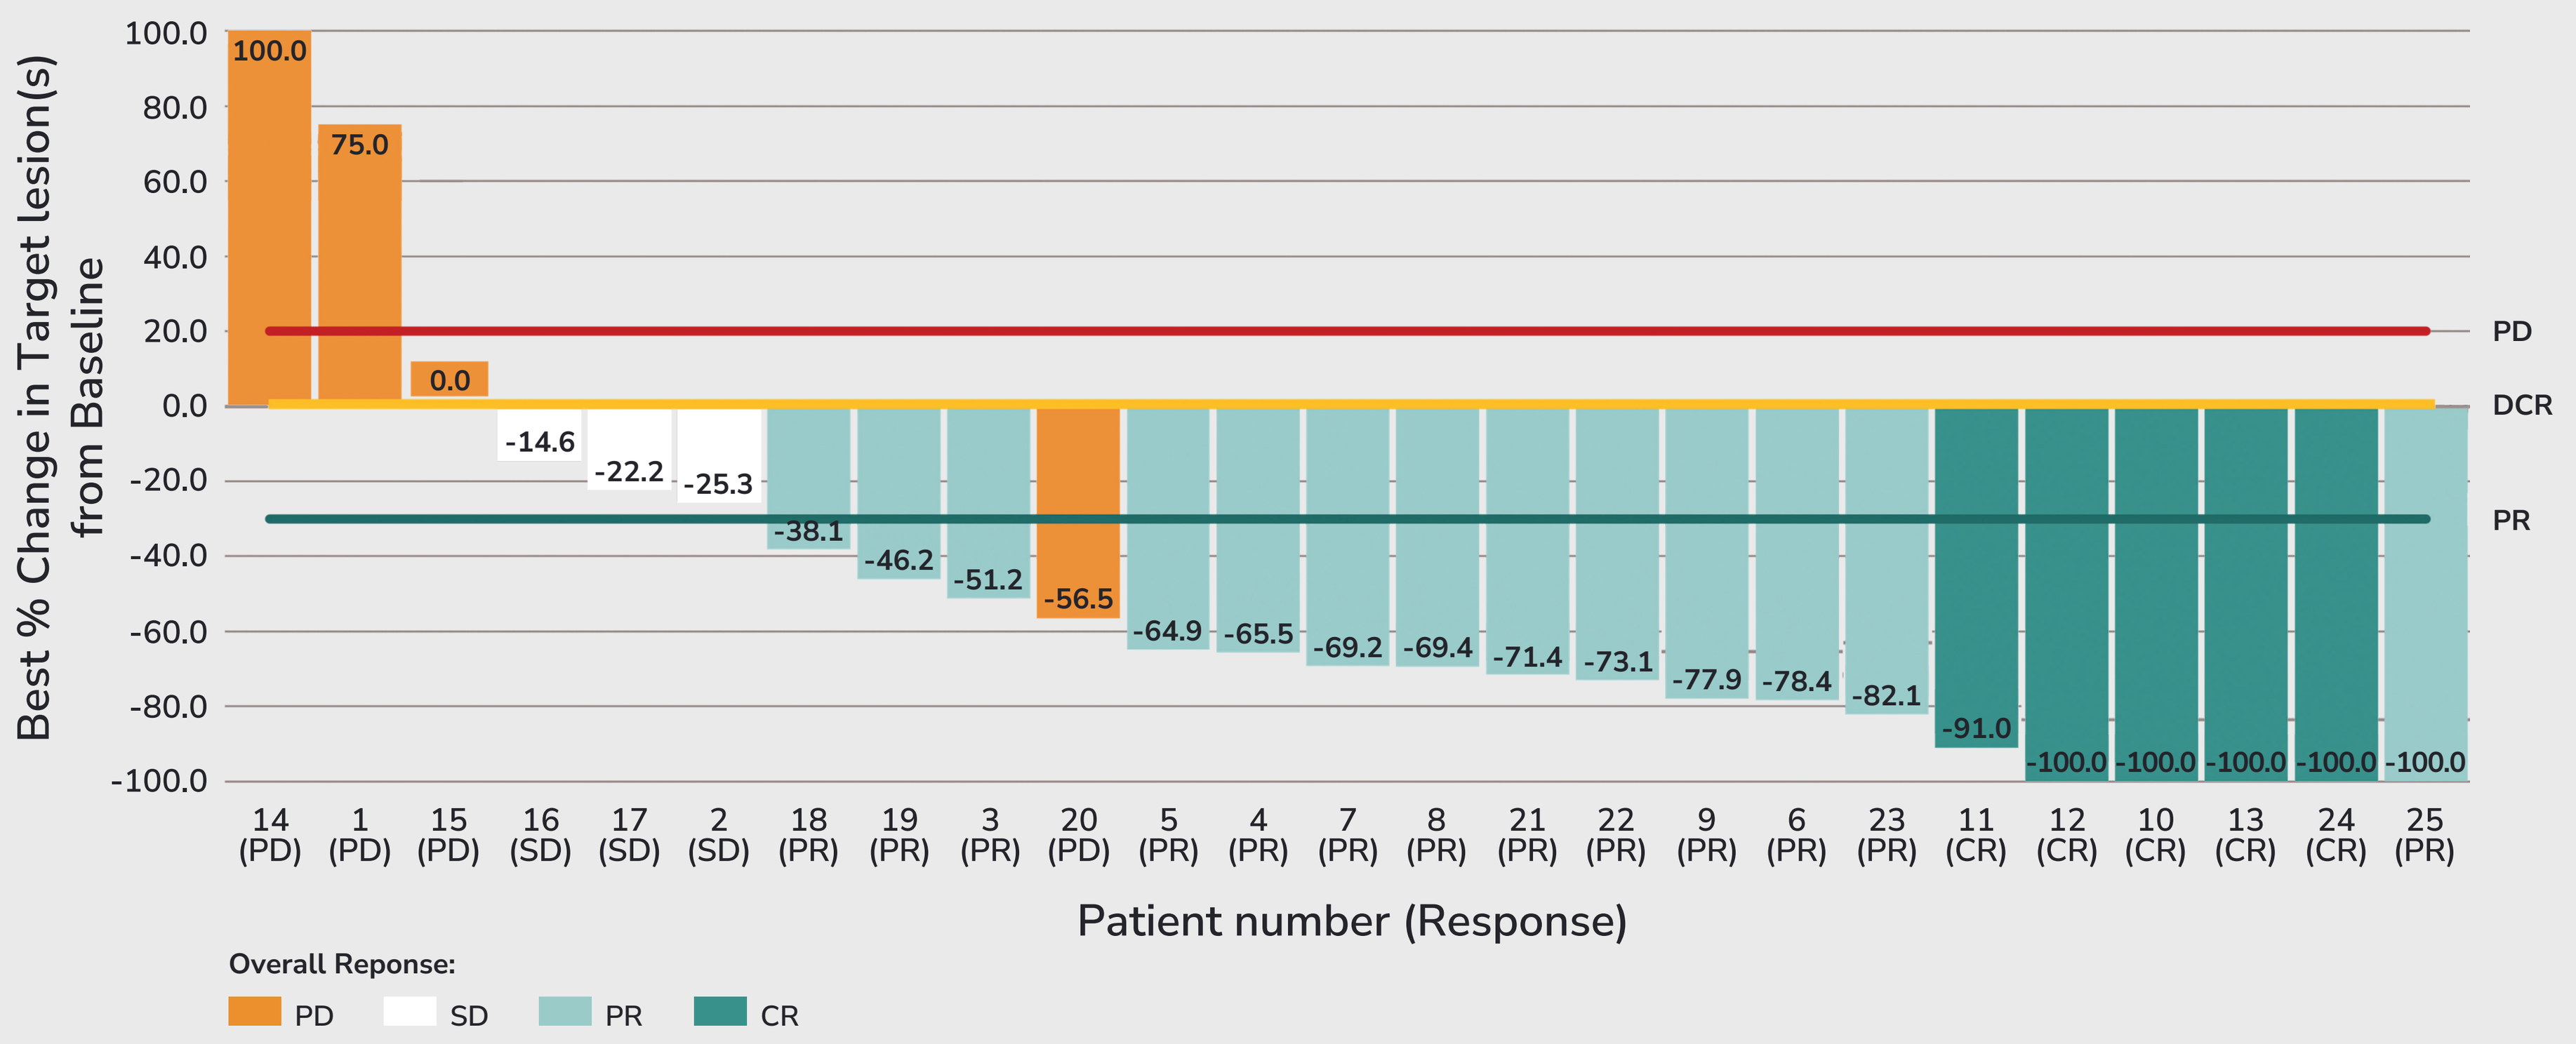
Best % Change in Target lesion(s) from Baseline
100.0
80.0
60.0
40.0
20.0
0.0
-20.0
-40.0
-60.0
-80.0
-100.0
PD
DCR
PR
100.0
75.0
0.0
-14.6
-22.2
-25.3
-38.1
-46.2
-51.2
-56.5
-64.9
-65.5
-69.2
-69.4
-71.4
-73.1
-77.9
-78.4
-82.1
-91.0
-100.0
-100.0
-100.0
-100.0
-100.0
14 (PD) 1 (PD) 15 (PD) 16 (SD) 17 (SD) 2 (SD) 18 (PR) 19 (PR) 3 (PR) 20 (PD) 5 (PR) 4 (PR) 7 (PR) 8 (PR) 21 (PR) 22 (PR) 9 (PR) 6 (PR) 23 (PR) 11 (CR) 12 (CR) 10 (CR) 13 (CR) 24 (CR) 25 (PR)
Patient number (Response)
Overall Reponse:
PD
SD
PR
CR

The waterfall plot demonstrates an ORR of 72% (18/25) with a disease control rate of 84% (21/25). Whilst the spider plots and progression free survival (PFS) curve confirms durability of the clinical response. The 80% PFS represents a 20% improvement in the PFS reported for ipilimumab plus nivolumab (CheckMate 067).

DURABILITY OF RESPONSE & SAFETY

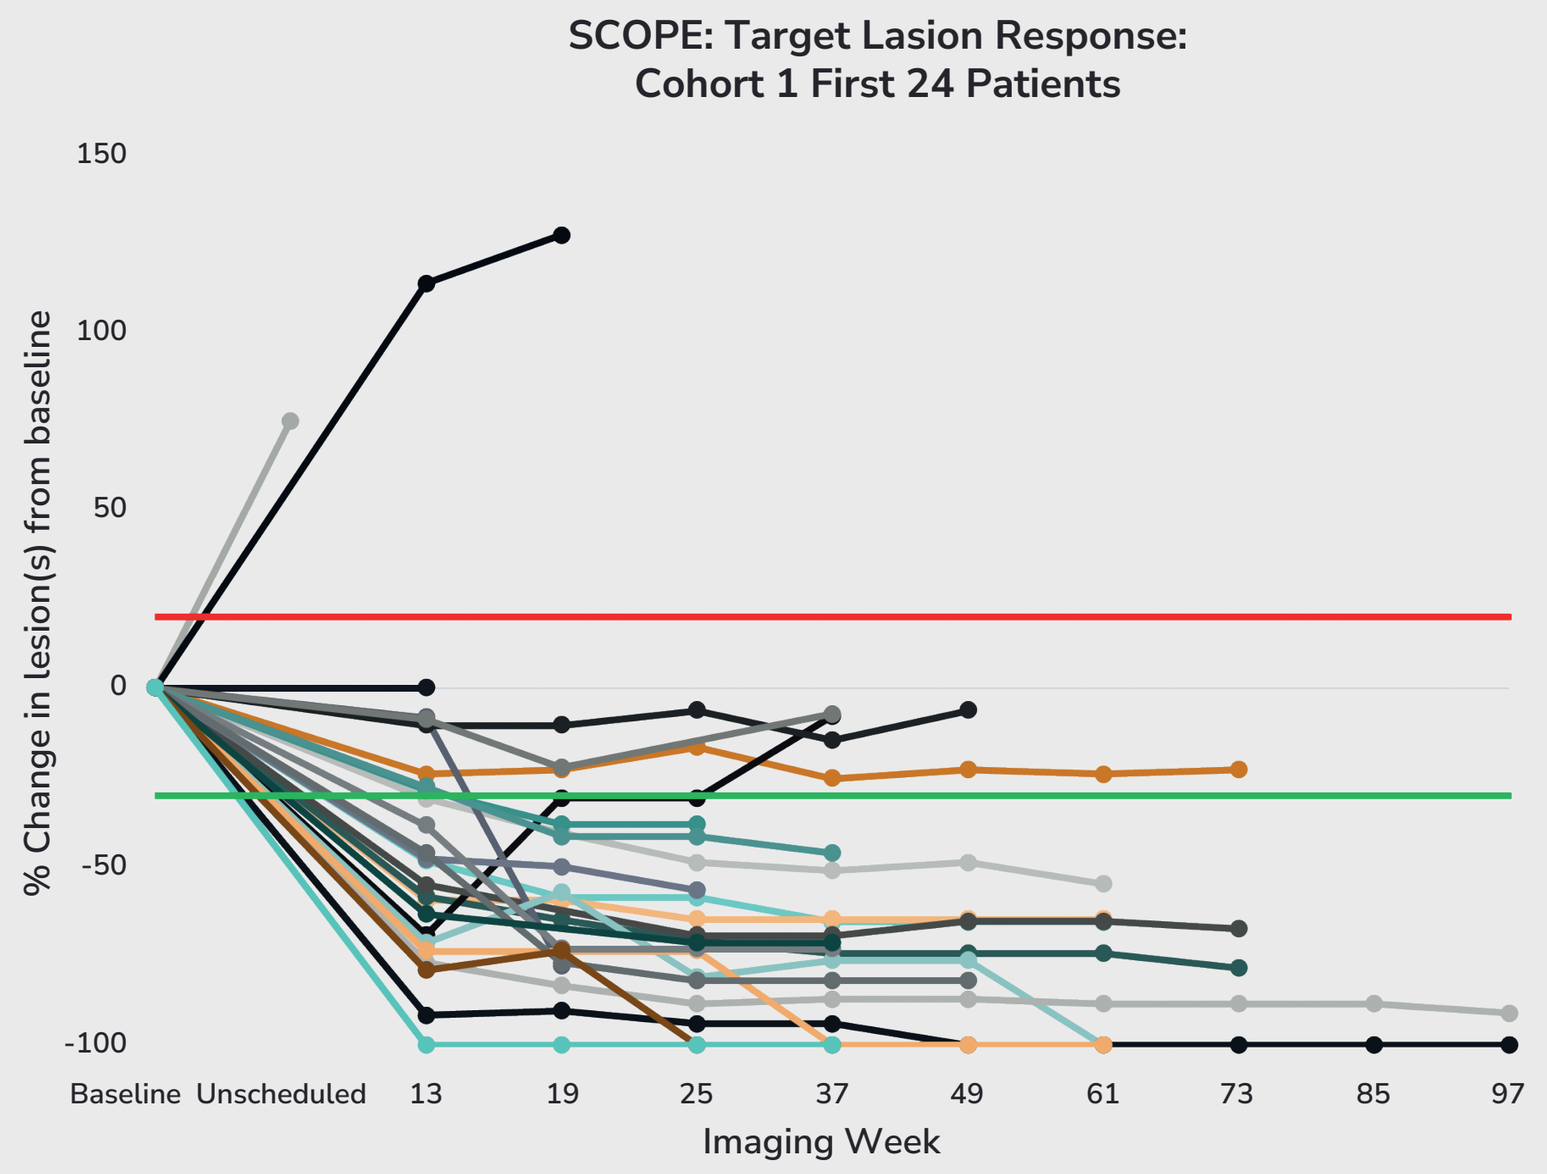
SCOPE: Target Lasion Response:
Cohort 1 First 24 Patients
% Change in lesion(s) from baseline
150
100
50
0
-50
-100
Baseline Unscheduled 13 19 25 37 49 61 73 85 97
Imaging Week

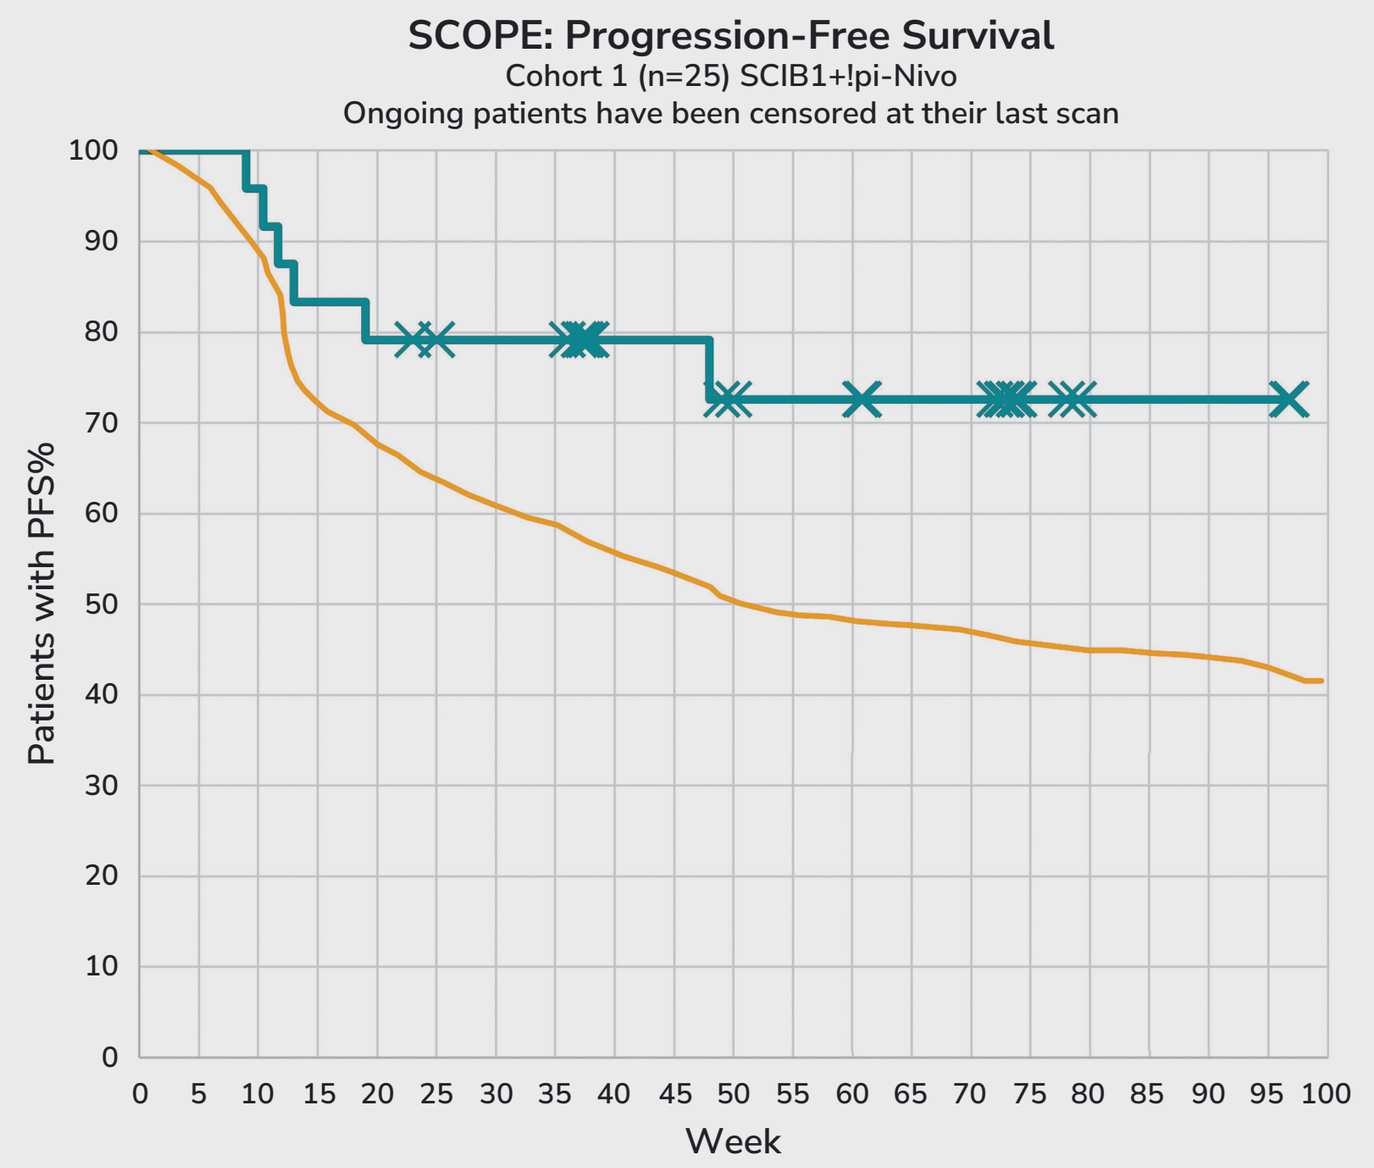
SCOPE: Progression-Free Survival
Cohort 1 (n=25) SCIB1+Ipi-Nivo
Ongoing patients have been censored at their last scan
Patients with PFS%
100
90
80
70
60
50
40
30
20
10
0
0 5 10 15 20 25 30 35 40 45 50 55 60 65 70 75 80 85 90 95 100
Week

Safety and tolerability was assessed by the recording of Adverse Events, physical examination, routine blood tests, injection site assessment, electrocardiogram & performance status. Most of the SCIB1-related adverse events were Grade 1/2. Four Grade 3 related adverse events were reported each in one patient: rash, neutropenia, pulmonary embolism and raised GGT. No exacerbation of immune-mediated adverse events was observed when SCIB1 was added to nivolumab with ipilimumab.

T-CELL RESPONSES

SCIB1-specific T cell responses were assessed using a cultured ELISpot assay following TRP-2 or gp100 peptides stimulation. The best response to any peptide at any time point post vaccination was compared with the matched baseline response. Wilcoxon matched-pairs signed rank test used to compare base line responses to post vaccine responses. It was possible to perform this test in 23 patients, of which 16 (70%) demonstrated a T-cell response, 12 (75%) of whom were responders and 15 (94%) had disease control.

BEST OVERALL RESPONSE	T CELL RESPONDERS	NON T CELL RESPONDERS	TOTAL (N=25)
CR	3	2	5
PR	9	2	13
SD	3	-	3
PD	1	3	4
Total (n=23)	**16 (70%)**	**7**	

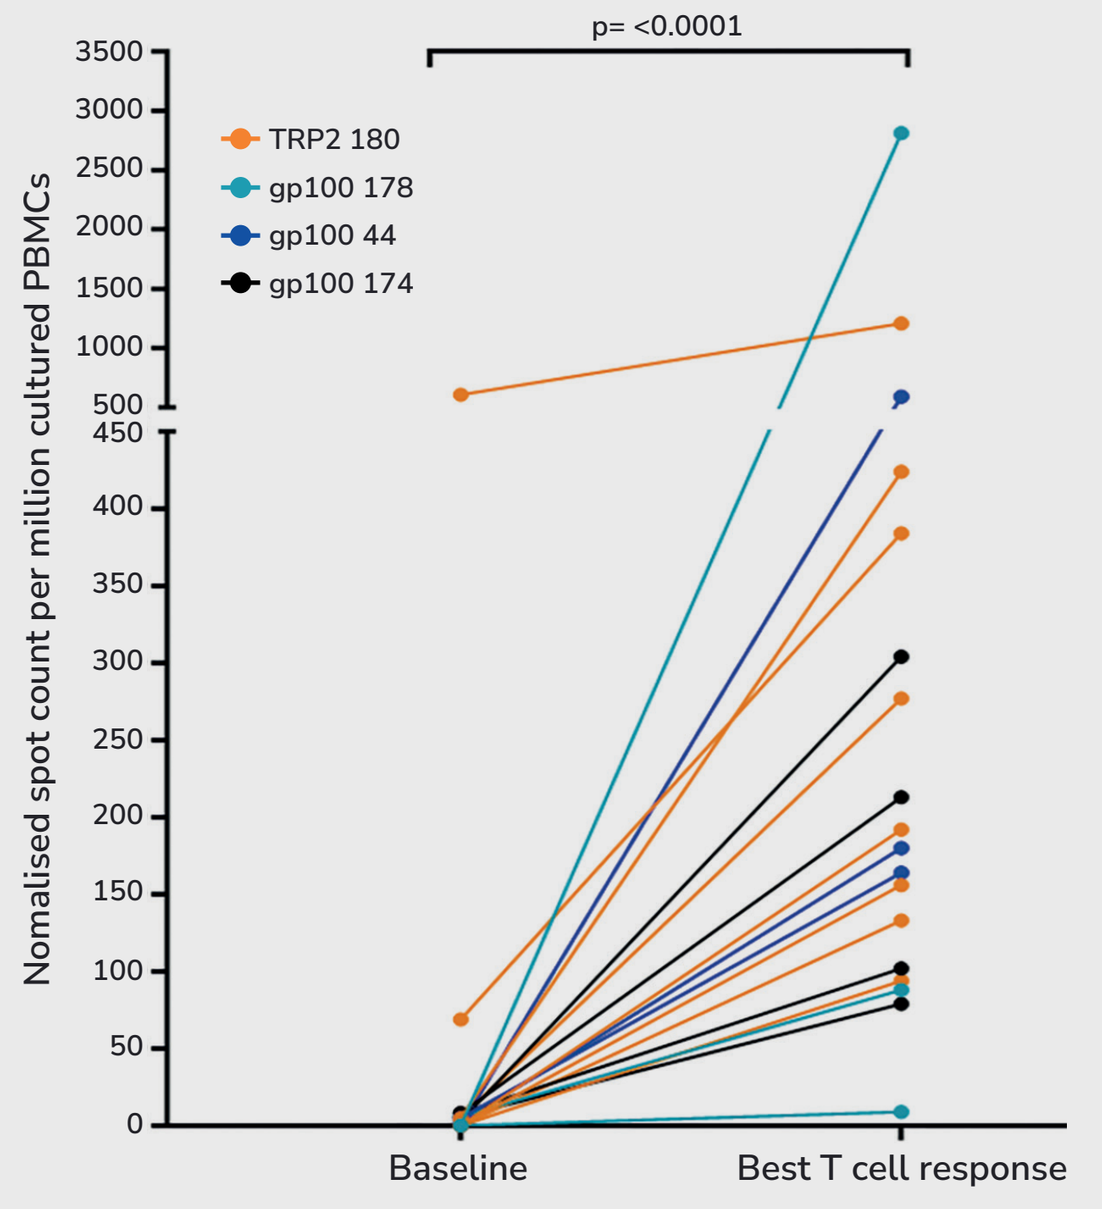
p= <0.0001
TRP2 180
gp100 178
gp100 44
gp100 174
Nomalised spot count per million cultured PBMCs
3500
3000
2500
2000
1500
1000
500
450
400
350
300
250
200
150
100
50
0
Baseline
Best T cell response

DISCUSSION & CONCLUSIONS

As per this preliminary analysis SCIB1 in combination with nivolumab and ipilimumab as first line treatment for unresectable Stage 3/4 melanoma resulted in 80% 6-month PFS with strong vaccine specific T cell responses in 70% of patients without an increase in clinically meaningful adverse events. The overall response rate was 72% and DCR was 84%. The study is now fully enrolled and continues with 43 patients. In addition, iSCIB1+, the next generation DNA melanoma vaccine based on Scancell's ImmunoBody technology administered by intra-muscular and intra-dermal needle free injection is being evaluated in a wider HLA patient population. The results from this group of studies encompassing over 140 patients will form the basis for the design of the follow-on global randomized control registration study in a larger patient population.

SCANCELL

CONTACT

Dr Nermeen Varawalla
Scancell Ltd

nermeenvarawalla@scancell.co.uk
scancell.co.uk

REFERENCES

1. Targeting gp100 and TRP-2 with a DNA vaccine: Incorporating T cell epitopes with a human IgG 1 antibody induces potent T cell responses that are associated with a favorable clinical outcome in a phase I/II trial. Patel et al, ONCOIMMUNOLOGY 2018, VOL,0, NO.0, e1433516
2. Evaluation of Two Dosing Regimens for Nivolumab in Combination With Ipilimumab in Patients With Advanced Melanoma: Results From the Phase IIIb/IV CheckMate 511 Trial. Lebber et al, J Clin Oncol. 2019 Apr 10;37(11):867-875. doi: 10.1200/JCO.18.01998